2051-Antitumor activity of a novel allogeneic colorectal cancer vaccine in C57BL/6 mice bearing MC38 anti-PD1 resistant colon carcinoma syngeneic model with TME

Céline Gongora[1], Elsa Kress[2], Claudine Vermot-Desroches[3], Lionel Chalus[4], Jeremy Forget[4], Benoît Pinteur[5], Paul Bravetti[5], Charles Dumontet[2], Antoine Italiano[6]

1.Institut de Recherche en Cancérologie de Montpellier, Montpellier, France; 2.Antineo, Lyon, France; 3. Therawings, Lyon, France; 4. BioElpida, Lyon, France; 5. Brenus Pharma, Issoire, France; 6. Institut Bergonié , Bordeaux, France

BACKGROUND

- Metastatic colorectal cancer (mCRC) is one of the major causes of death worldwide.
- The relative therapeutic resistance of these populations to I/O drives the need for new treatment.
- STC-1010 (Brenus Pharma) therapeutic vaccine is developed by tumor cells stimulation to induce overexpression of TAA and neoantigens to mimic mCRC resistant cancer cells allowing education of the immune system to target the patient's tumor cells harboring the same resistance factors.

- We report results of a study evaluating efficacy and safety of a 3 cell lines-based product namely "3CL-SH" (made of CT26, CMT93 and LTPA) physicaly stimulated (S=irradiation and heat shock) and haptenized (H) with immunostimulant in an anti-PD1 resistant MC38 colorectal tumour model grafted with its TME.

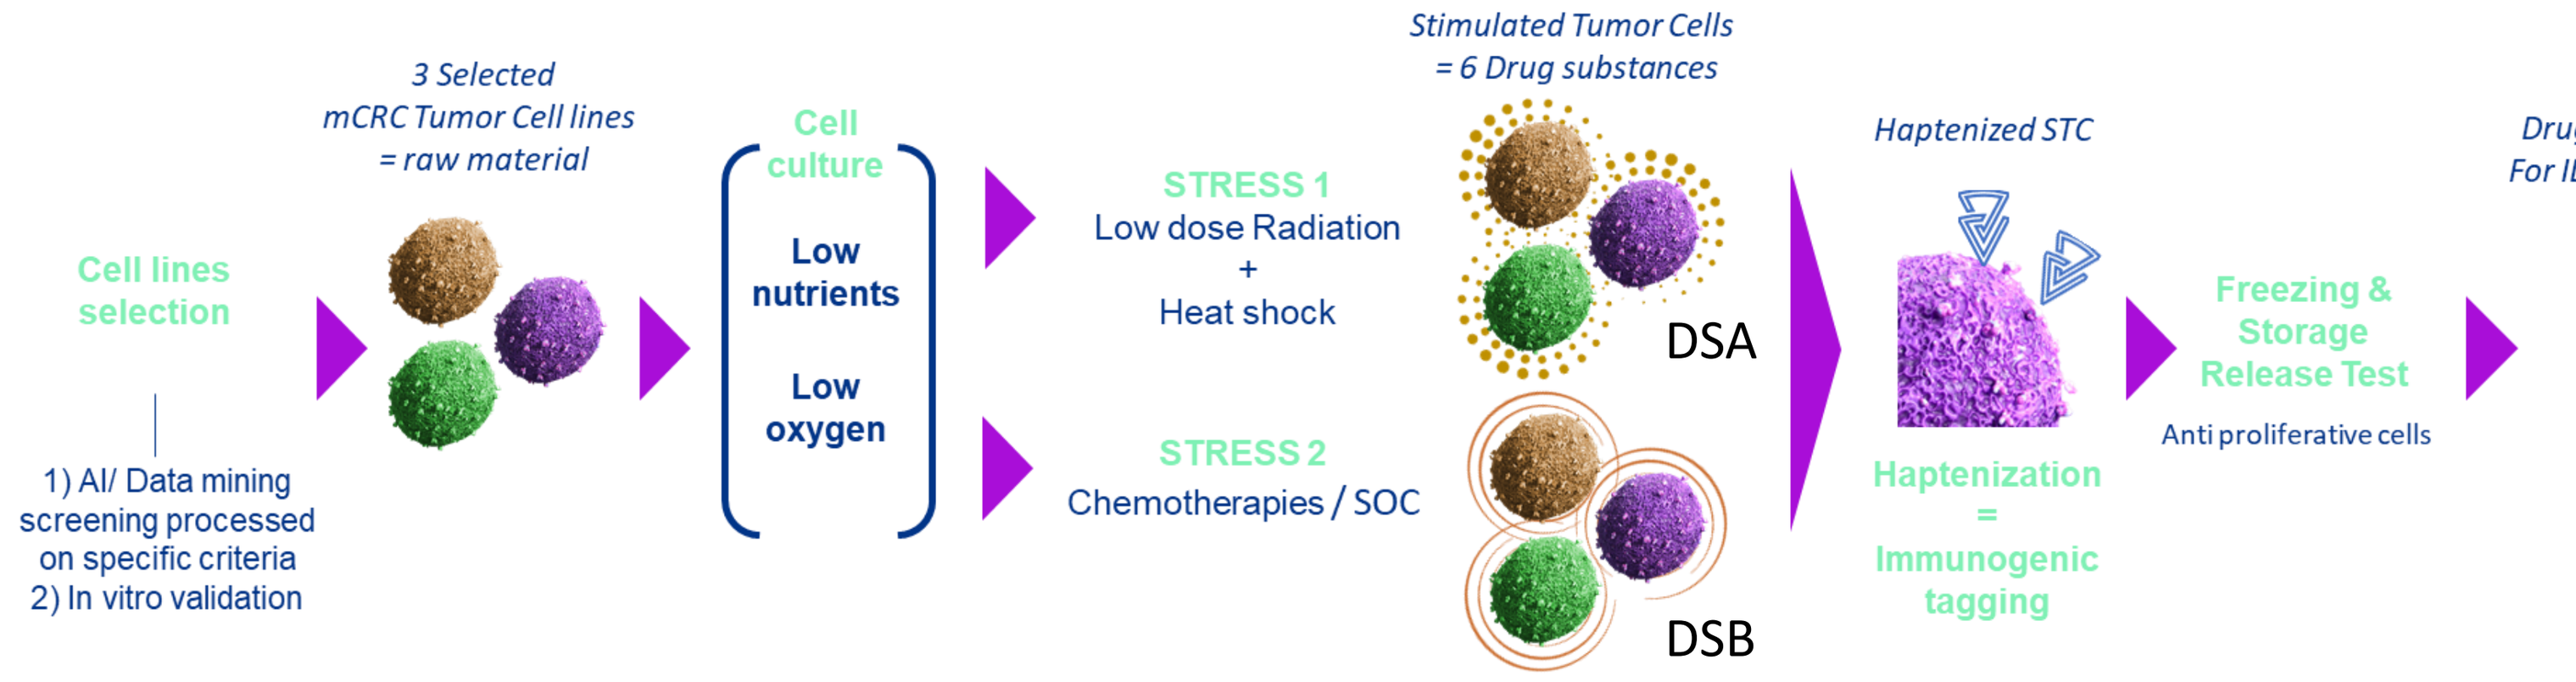
Cell lines selection
1) AI/ Data mining screening processed on specific criteria
2) In vitro validation
3 Selected mCRC Tumor Cell lines = raw material
Cell culture
Low nutrients
Low oxygen
STRESS 1
Low dose Radiation + Heat shock
STRESS 2
Chemotherapies / SOC
Stimulated Tumor Cells = 6 Drug substances
DSA
DSB
Haptenized STC
Haptenization = Immunogenic tagging
Freezing & Storage Release Test
Anti proliferative cells
Drug Product For ID injection

Fig 1. STC (Stimulated Tumor Cells) Technology

METHODS

- Female C57BL/6 mice bearing MC38 tumour model grown in vivo under anti-PD1 selection pressure (anti-PD1; 12.5 mg/kg; qw) to obtain MC38 resistant to anti-PD1 treatment (MC38 antiPD1R). Two sampled tumors from these mice were cut into fragment of 2 mm^2 and reimplanted in the right flank of naïve syngenic mice.
- 5 groups (15 mice per group) were allocated to:

 G1) Control group: Isotype/placebo/vehicle
 G2) Anti-PD1/placebo/vehicule
 G3) Anti PD1/placebo-GM-CSF/endoxan
 G4) Isotype/3CL-SH-GM-CSF/endoxan
 G5) Anti-PD1/3CL-SH-GM-CSF/endoxan.

- Endoxan, vehicule and antibodies were administered by IP, vaccine or placebo subcutaneously.
- Overall survival (OS) and tumour growth (TG) were recorded until 1600 mm3 or safety endpoint.
- 5 mice per group were euthanized and sampled for immunophenotyping.
- In parallel, we conducted the identification and relative quantification of proteins expressed in 3CL-SH by LC-MS/MS and flow cytometry.

RESULTS

- Flow cytometry analysis shows that HSP-70 expression was significantly upregulated in each of the three cell lines of the 3CL-SH compared to untreated cells.
- LC-MS/MS analysis demonstrated that cell lines of 3CL-SH display overexpression of membrane proteins of interest such as Lamp 1, MDR1A/B and Fas Receptor.
- The group treated with 3CL-SH significantly improves the survival of mice bearing MC38PD1R tumor model vs the control group (Log-rank Mantel-Cox test, pvalue=0.0368) and we observed a non-benefit of the combination of 3CL-SH and anti-PD1 drug in concomitant injections.
- Immunophenotyping data show statistical increase in the infiltration of anti-tumour populations (CD8+ T cells, TAM1) in the tumours treated with 3CL-SH but also an increase in M2 that could be linked with an excessive dosage of GM-CSF.
- No side effect or inflammatory reaction towards the 3CL-SH is evidenced.

HSP-70 expression by flow cytometry

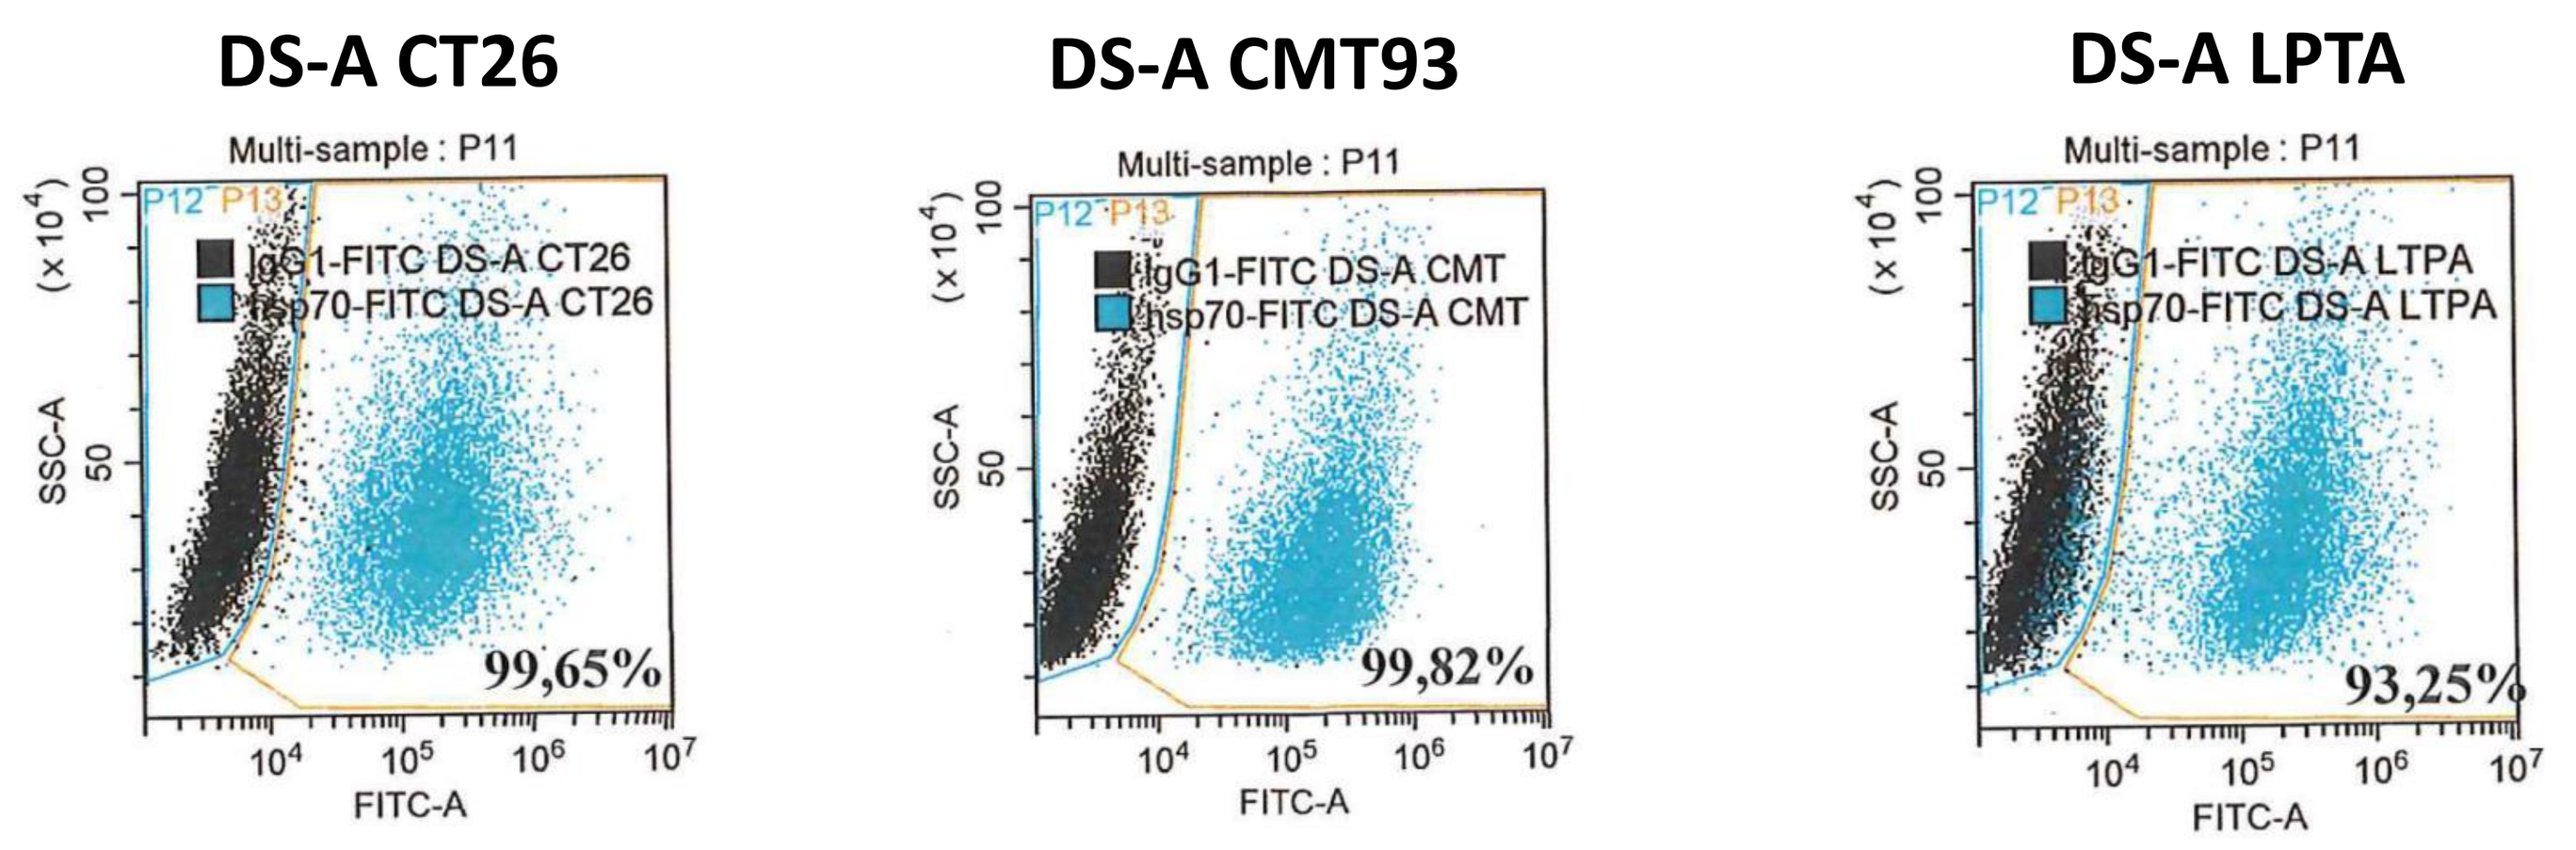
DS-A CT26
DS-A CMT93
DS-A LPTA
Multi-sample : P11
IgG1-FITC DS-A CT26
hsp70-FITC DS-A CT26
IgG1-FITC DS-A CMT
hsp70-FITC DS-A CMT
IgG1-FITC DS-A LTPA
hsp70-FITC DS-A LTPA
SSC-A
FITC-A
99,65%
99,82%
93,25%

Fig 2. HSP-70 expression by flow cytometry analysis for DSA CT26, DSA CMT93, DSA LPTA

LC-MS/MS Proteomic Analysis results

Cell model/marker	Fas Receptor	Lamp1	MDR1A	MDR1B
CMT93	DSA+ DSB+	ND		
CT26	DSB +	DSA-		DSA-
LTPA	DSA +	DSA+ DSB+	DSA+	DSA+ DSB+

*ND: no differential, DSA+/-: overexpressed or underexpressed after physical stress;
DSB+: overexpressed after chemical stress

Fig 3. LC-MS/MC Results focus on interesting markers depending on cell lines used for the 3CL-SH

Tumor volumes (mm3), all compounds at D14

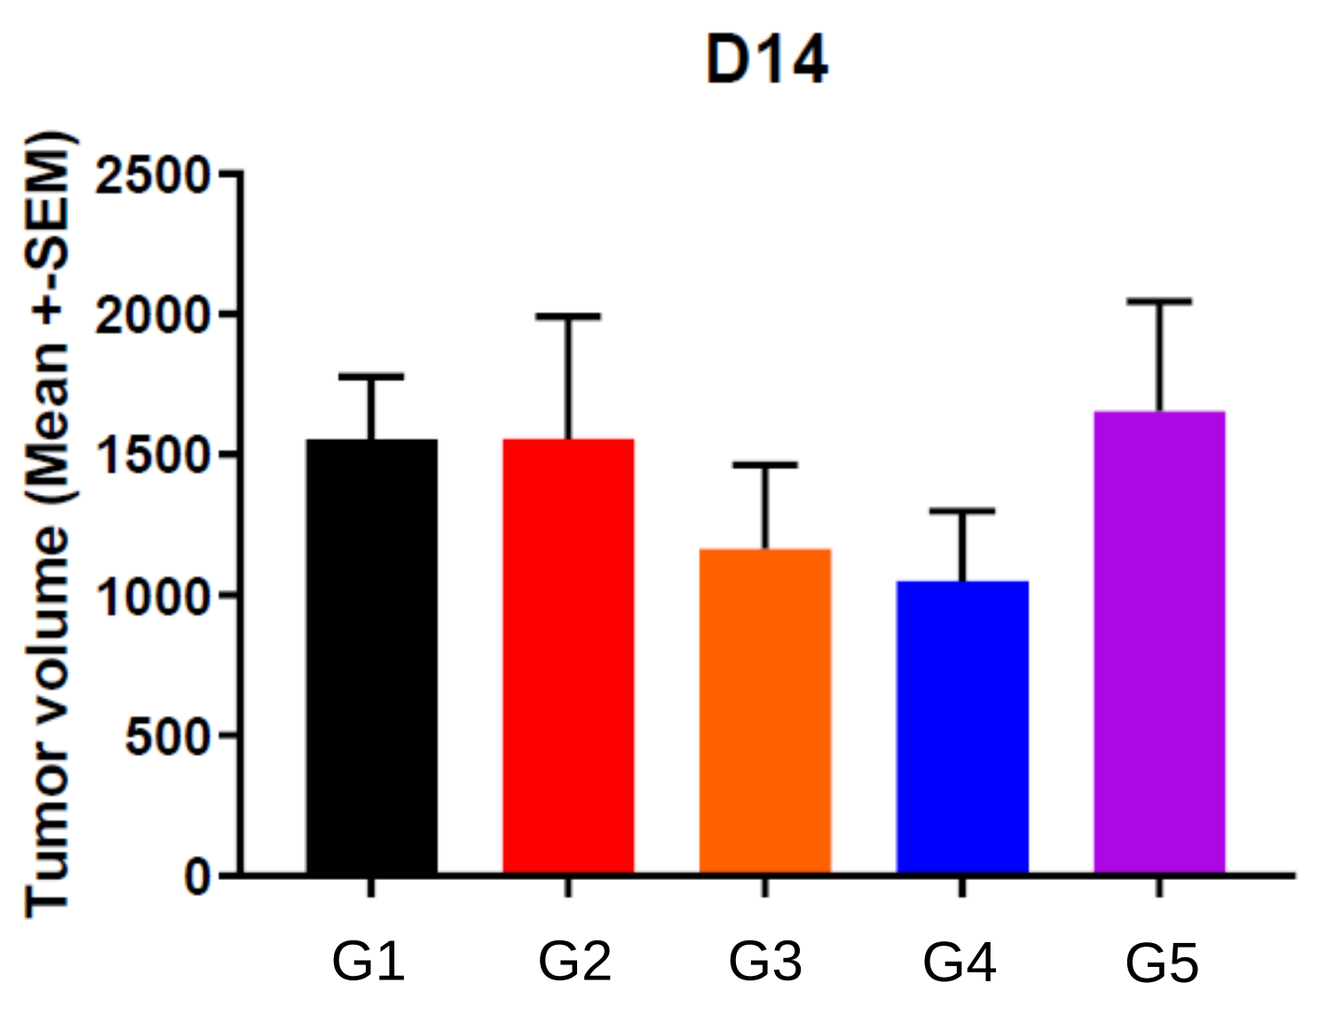
D14
Tumor volume (Mean +-SEM)
G1
G2
G3
G4
G5

Fig 4. Tumor volume (mean + SEM) measured by Caliper tested on PD1R tumor growth

Individual Tumor volumes (mm3) for 3CL-SH-GM-CSF+Endoxan group

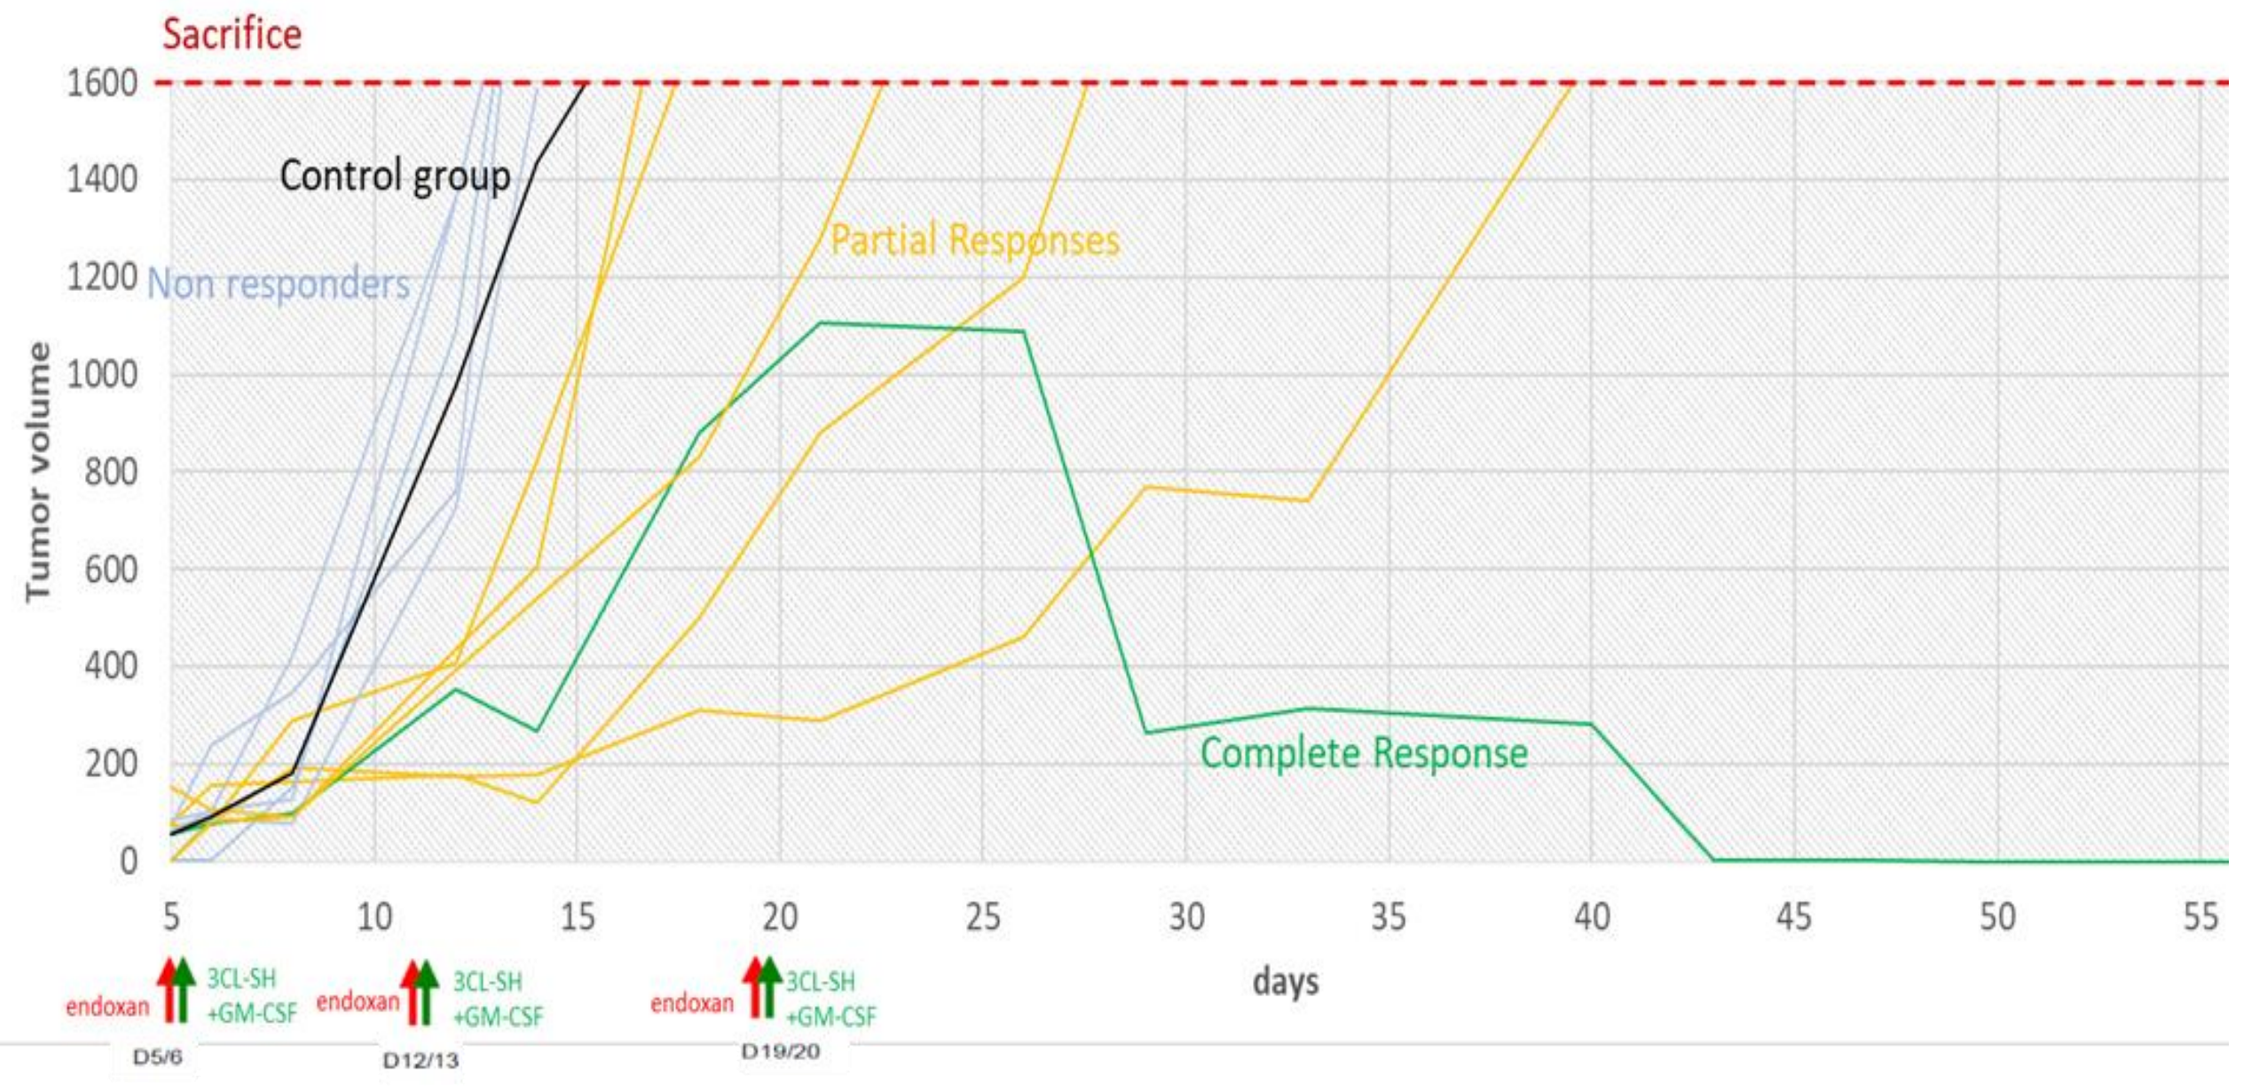
Sacrifice
Control group
Non responders
Partial Responses
Complete Response
Tumor volume
days
endoxan
3CL-SH +GM-CSF

Fig 5. Individual curves of Tumor growth for group 4 vs median control group

Overall Survival (days) for 3CL-SH-GM-CSF+Endoxan group

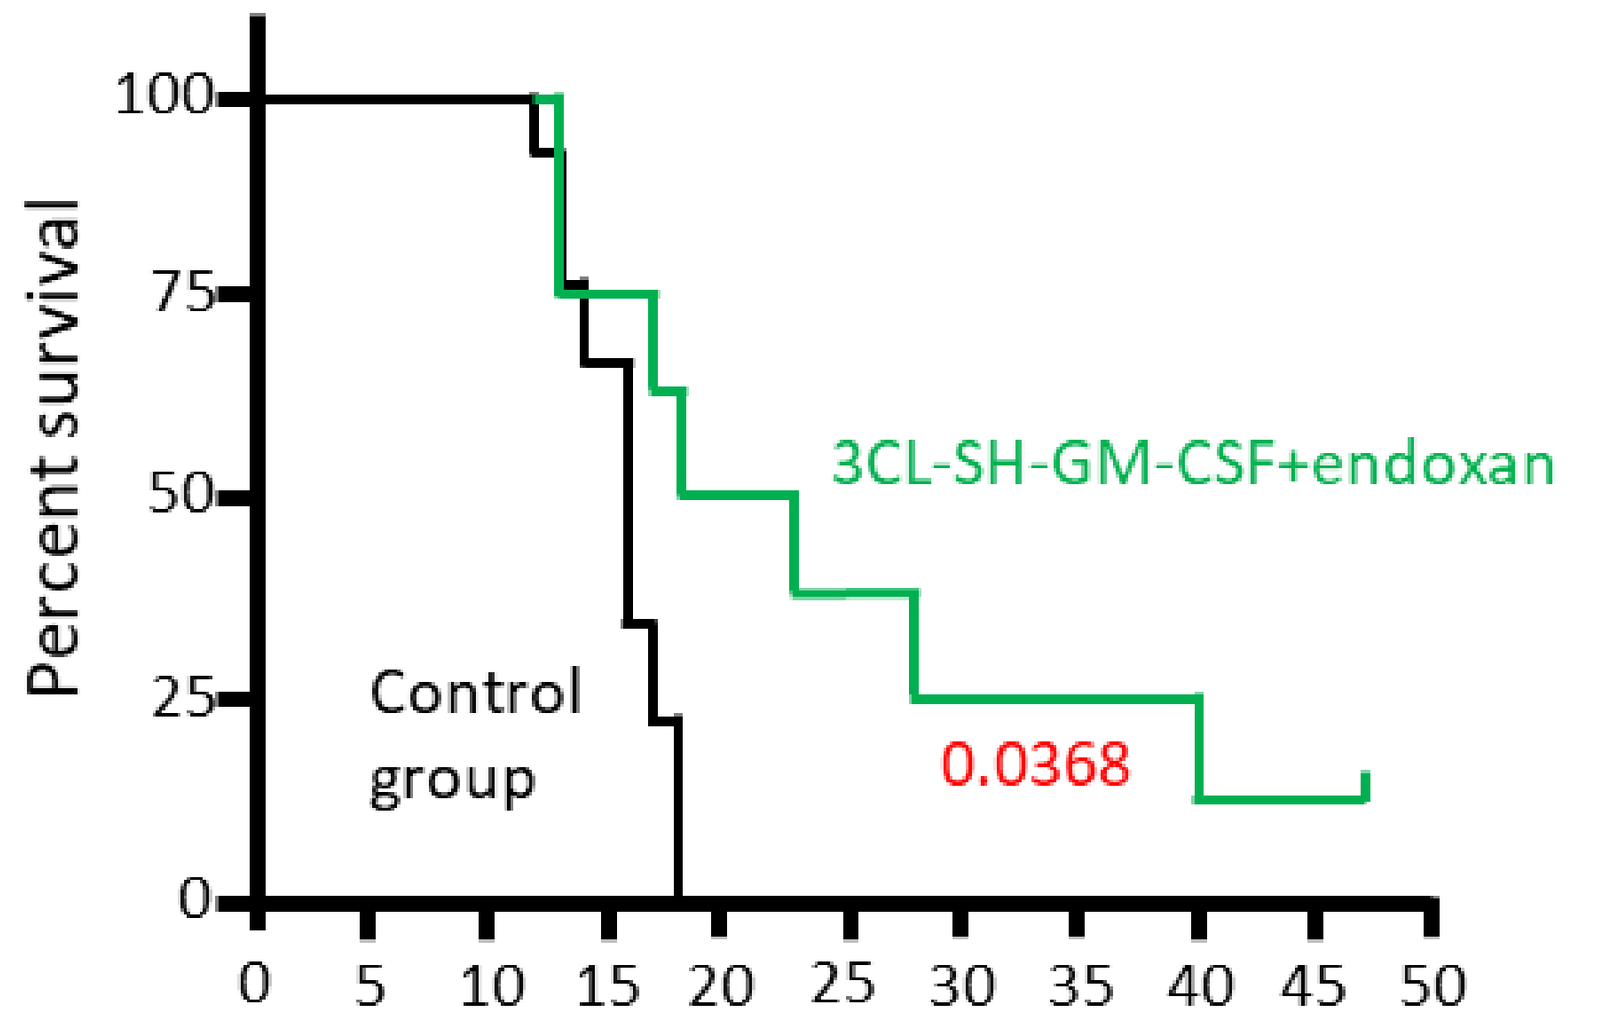
Percent survival
3CL-SH-GM-CSF+endoxan
Control group
0.0368

Fig 6. Overall Survival for group 4 vs control group (Log-rank Mantel-Cox test)

Immunophenotyping analysis (n=25)

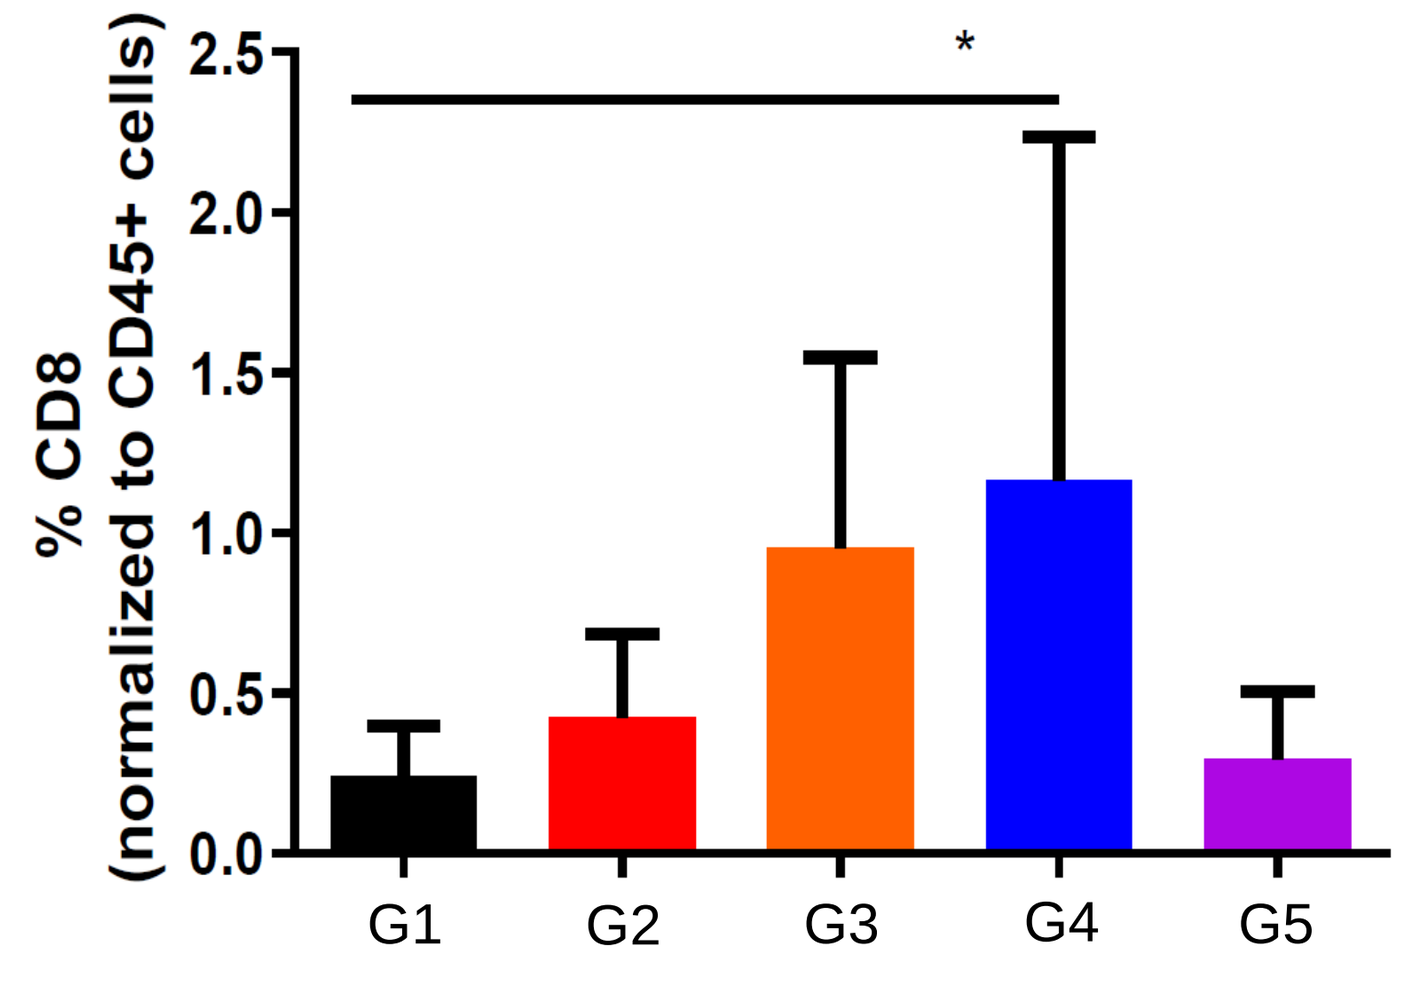
% CD8 (normalized to CD45+ cells)
*
G1
G2
G3
G4
G5

Fig 7. CD8+ infiltration per group at D14

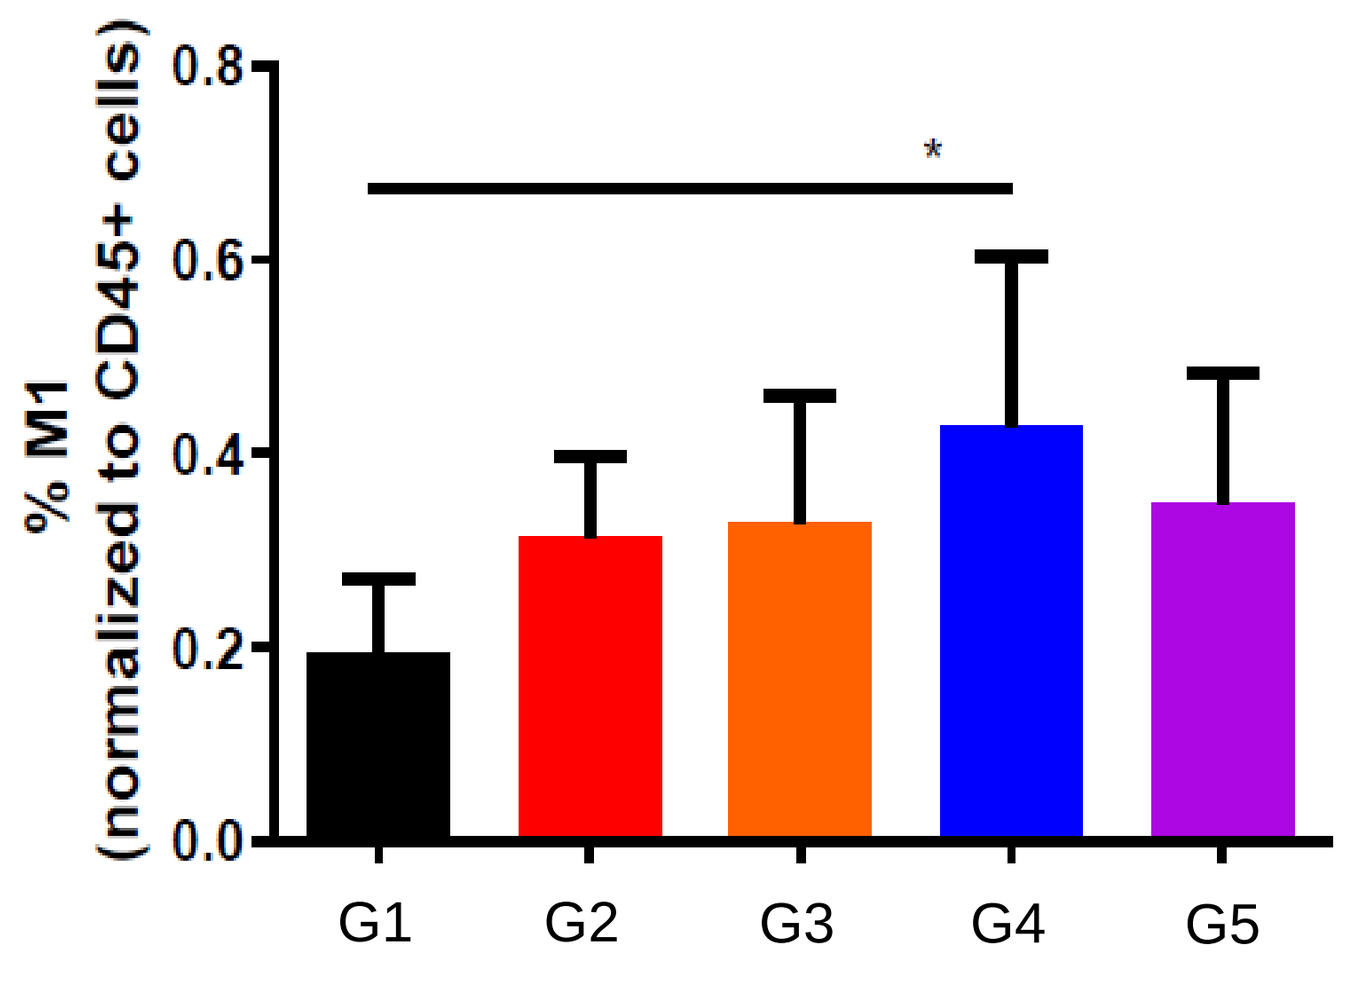
% M1 (normalized to CD45+ cells)
*
G1
G2
G3
G4
G5

Fig 8. TAM1 infiltration in the TME at D14

CONCLUSIONS

- The 3CL-SH treatment presents a promising antitumor efficacy on the aggressive MC38 PD1R model, resulting in a significative gain of survival and anti-tumoral changes in the immune infiltrate of treated animals.
- More studies are needed to evaluate the benefit of a combination with ICI.

Contact : Céline, Gongora, Ph. D.,
celine.gongora@inserm.fr

Acknowledgment : Anaquant, Lyon, France